Characterization of JAK2 V617F Allele Burden in Advanced Myelofibrosis (MF) Patients: No Change in V617F:WT JAK2 Ratio in Patients with High Allele Burdens Despite Profound Clinical Improvement Following Treatment with the JAK Inhibitor INCB018424

S. Verstovsek,[1] H. M. Kantarjian,[1] A. D. Pardanani,[2] T. Burn,[3] K. Vaddi,[3] J. Redman,[3] E. Bradley,[3] R. Levy,[3] S. Friedman,[3] G. Hollis,[3] and A. Tefferi[2]

[1]*University of Texas M. D. Anderson Cancer Center, Houston, Texas,* [2]*Mayo Clinic, Rochester, Minnesota, and* [3]*Incyte Corporation, Wilmington, Delaware, USA*

1 Abstract

Background: Activating mutations in the pseudokinase domain of JAK2 occur at a high frequency in Philadelphia chromosome-negative myeloproliferative disorders (MPDs). Increasing JAK2 V617F allele burden has been shown to correlate with disease severity (bone marrow dysfunction, organomegaly and constitutional symptoms), which is consistent with exaggerated JAK2 signaling playing a central role in MPDs. INCB018424 is a potent and selective JAK1/2 inhibitor currently being evaluated in the clinic for the treatment of MPDs including primary myelofibrosis (PMF), post-polycythemia vera, and essential thrombocythemia myelofibrosis (post-PV/ET MF).

Methods: The percentage of mutant JAK2 V617F relative to wild-type JAK2 was determined in peripheral blood using a quantitative single-nucleotide extension assay. The assay is based on the single nucleotide extension of a DNA primer that anneals adjacent to the V617F mutation. The percentage of JAK2 V617F in bone marrow was determined using pyrosequencing, as previously described. The relationship between JAK2 V617F allele burden and clinical phenotype was assessed with samples obtained at study entry. JAK2 V617F allele burden was assessed in nineteen PMF and post-PV/ET MF patients treated with INCB18424 25 mg BID for a minimum of 3 months using peripheral blood and bone marrow.

Results: There is excellent correlation in the JAK2 V617F allele burden between samples obtained from bone marrow and peripheral blood, suggesting that the fraction of cells bearing the mutant clone remains stable during hematopoiesis in this patient group. The mean %V617F values were 59%, 80% and 86% for post-ET MF (n=2), post-PV MF (n=8), and PMF (n=9) patients. V617F allele burden correlated with spleen size, absolute neutrophil count (ANC), and body mass index (BMI) in this patient population (R^2 values of 0.61, 0.17 and 0.44, respectively), with a higher allele burden predicting larger spleen size, a higher ANC, and a lower BMI. Eighteen of the 19 evaluable JAK2 V617F-positive patients had baseline JAK2 V617F values above 50%, with the mean %V617F value for all evaluable patients being 81%, implying that the majority of hematopoietic stem cells contain the mutant allele in this patient population. During INCB18424 therapy (at least 3 months of treatment) in these patients, profound improvements in clinical endpoints including reduction or resolution of splenomegaly and constitutional symptoms are observed, yet only a modest decrease in JAK2 V617F allele burden is seen in both bone marrow and peripheral blood (13% and 9% decrease from baseline, respectively, at 3 months).

Conclusions: While the patient group is small and heterogeneous in nature, clinical correlates of V617F allele burden observed in MF patients in this study are similar to those reported in PV. In addition, despite profound clinical improvements in these patients, V617F allele burdens do not change dramatically, suggesting that the clinical activity of INCB018424 might involve inhibition of downstream signaling in cells harboring activating JAK2 mutations rather than changing the allele burden.

2 Background

- Activating mutations of JAK2 occur at a high frequency in Philadelphia chromosome-negative MPDs
- JAK2 V617F is the predominant activating mutation of JAK2
- In MF patients, high JAK2 V617F allele burden is associated with advanced disease and clinical signs and symptoms
 - Splenomegaly
 - Constitutional symptoms (eg. Fatigue, pruritis)
 - Leukocytosis
- INCB018424 is a potent JAK1/2 inhibitor, with > 80-fold selectivity against JAK3, that has been shown to potently inhibit signaling of cytokines that are elevated in MF patients
- Treatment of MF patients with INCB018424 results in profound clinical improvement characterized by:
 - reduction in spleen size (see Verstovsek et al., ASH Abstract 1762)
 - reduction in inflammatory cytokines (see Tefferi et al., ASH Abstract 2804)
 - improvement in constitutional symptoms (see Verstovsek et al., ASH Abstract 1762)
 - Improvement in nutritional status (see Mesa et al., ASH Abstract 1760)

3 Objectives

- Evaluate JAK2 V617F allele burden in primary MF (PMF) and post-PV/ET MF patients enrolled in study INCB18424-251
 - Compare peripheral blood and bone marrow JAK2 V617F allele burdens
 - Correlate clinical signs and symptoms of disease with baseline JAK2 V617F allele burdens
- Evaluate changes in JAK2 V617F allele burden over time with INCB018424 treatment in MF
- Evaluate the relationship between changes in V617F allele burden and the magnitude of clinical improvement in MF patients treated with INCB018424

4 INCB018424 Mechanism of Action

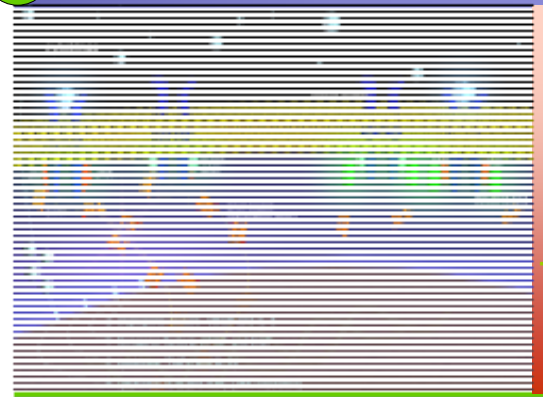

- INCB018424, a JAK1/2 inhibitor, blocks cytokine signaling and amplified cytokine and growth factor expression

5 Methods

- JAK2 V617F levels were evaluated in V617F-positive MF patients treated with INCB018424 25 mg BID for a minimum of 3 months
 - JAK/STAT activating mutations other than JAK2 V617F were not assessed
- JAK2 V617F allele burden:
 - The percentage of JAK2 V617F (relative to total JAK2) was determined by quantitative single-nucleotide extension and/or real-time PCR for peripheral blood and pyrosequencing for bone marrow
 - JAK2 V617F allele burden was assessed at study entry and on Day 1 of each 28 day cycle for Cycles 2-4 and every three cycles there after
- The relationship between baseline JAK2 V617F allele burden and clinical signs and symptoms was assessed
- The relationship between changes in JAK2 V617F allele burden on study and clinical signs and symptoms was assessed
 - Cytokines were assessed by non-biased proteome analysis (see Tefferi et al., ASH Abstract 2804)
 - Patient symptoms were assessed using MFSAS (see Mesa et al., ASH Abstract 1760)

6 INCB18424-251: JAK2 V617F Allele Burden in Bone Marrow and Peripheral Blood

- There is excellent correlation between bone marrow and peripheral blood V617F values in study INCB18424-251
- V617F levels in peripheral blood samples are an excellent surrogate for V617F allele burden in bone marrow in MF patients in this study

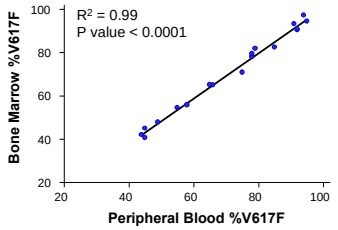
R² = 0.99
P value < 0.0001
Bone Marrow %V617F
Peripheral Blood %V617F

7 INCB18424-251: Baseline JAK2 V617F Allele Burden in MF Patients

- The majority of MF patients in study INCB18424-251 have high baseline JAK2 V617F allele burdens
- Average %V617F for V617F-positive patients prior to therapy was 75%
 - Average V617F allele burden in the MF patients in the current study is higher than the allele burdens in PV (45%) or ET (26%) reported by Vanucchi et al. (2008)
 - V617F allele burdens > 50% indicate the presence of homozygous V617F cells
- These data suggest that MF patients on this study have advanced disease

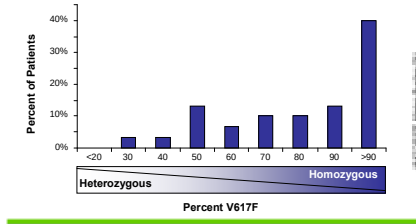
Percent of Patients
<20
30
40
50
60
70
80
90
>90
Heterozygous
Homozygous
Percent V617F

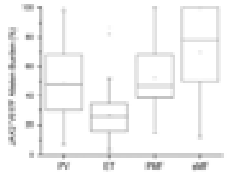

Source: Vanucchi et al 2008

8 INCB18424-251: Distribution of Baseline V617F Allele Burden by MF Subgroups

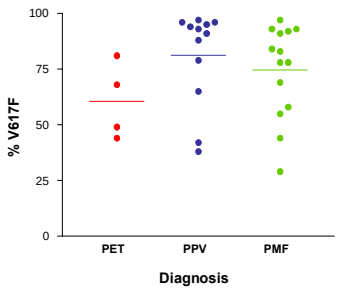
% V617F
PET
PPV
PMF
Diagnosis

- Mean baseline %V617F values were 61, 81 and 75% for post-ET (PET), post-PV (PPV) and PMF patients, respectively

9 INCB18424-251: Baseline JAK2 V617F Allele Burden and Splenomegaly

- In study INCB18424-251, JAK2 V617F allele burden correlates with splenomegaly
- A similar relationship between V617F allele burden and spleen size has been described by Vanucchi et al. (2008)

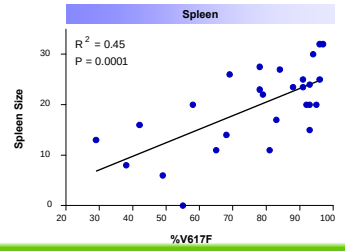
Spleen
R² = 0.45
P = 0.0001
Spleen Size
%V617F

10 INCB18424-251: Baseline JAK2 V617F Allele Burden and White Cell Counts

- In study INCB18424-251, baseline JAK2 V617F allele burden correlates with white cell counts, also as reported by Vanucchi et al. (2008)

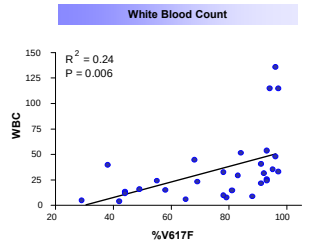
White Blood Count
R² = 0.24
P = 0.006
WBC
%V617F

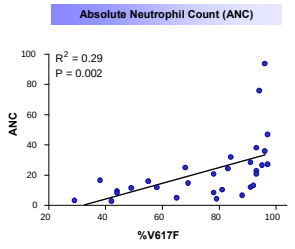
Absolute Neutrophil Count (ANC)
R² = 0.29
P = 0.002
ANC
%V617F

11 INCB18424-251: Baseline JAK2 V617F Allele Burden and Nutritional Status

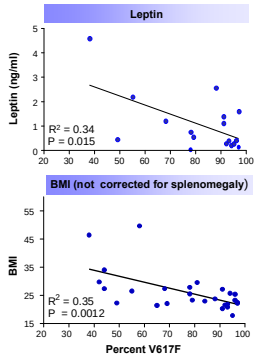
Leptin
Leptin (ng/ml)
R² = 0.34
P = 0.015
BMI (not corrected for splenomegaly)
BMI
R² = 0.35
P = 0.0012
Percent V617F

- In study INCB18424-251, baseline JAK2 V617F allele burden inversely correlates with markers of nutritional status
- Leptin is an adipose tissue derived hormone and plasma leptin levels are proportional to fat mass (Wallace et al., 1998)
 - Average leptin levels in healthy volunteers are 18.7 ng/ml for women (BMI range 23-36) and 6.5 ng/ml for men (BMI range 24-34)
- Mean baseline value of MF patients included in this study is 1.1 ± 1.2 ng/ml
 - Indicates adipose tissue wasting in study INCB18424-251 MF patients (Mesa et al., ASH Abstract 1760)

12 Changes in V617F Allele Burden in Study INCB18424-251

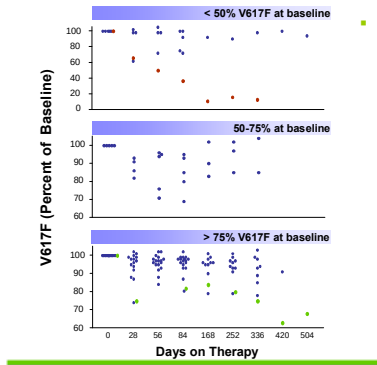
< 50% V617F at baseline
50-75% at baseline
> 75% V617F at baseline
V617F (Percent of Baseline)
Days on Therapy

- Modest but statistically significant decreases in JAK2 V617F are observed during INCB018424 therapy
 - A PPV-MF patient with one of the lower baseline V617F allele burdens has had a 90% decrease in V617F allele burden (highlighted in red)
 - PPV-MF patient with longest time on therapy has had a 32% decrease in V617F allele burden (highlighted in green)

13 Clinical Responses in Study INCB18424-251

- Similar clinical responses are observed following INCB018424 therapy, independent of the magnitude of V617F allele burden decrease

Decrease in spleen size on Day 84

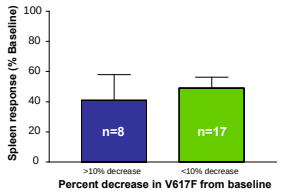
Spleen response (% Baseline)
n=8
n=17
>10% decrease
<10% decrease
Percent decrease in V617F from baseline

Decrease in inflammatory cytokines: Baseline vs Day 28

	V617F: decrease from baseline	
	>10% (n=6)	<10% (n=11)
CRP	-93%	-87%
IL-1β	-89%	-49%
IL-1RA	-81%	-68%
IL-6	-69%	-64%
IL-8	-72%	-74%
TNF	-56%	-32%
TNFRII	-75%	-50%

Differences between groups not statistically significant.

14 Clinical Responses in Study INCB18424-251

- Profound clinical improvement is seen in JAK2 V617-positive and in JAK2 V617-negative MF patients

Decrease in spleen size

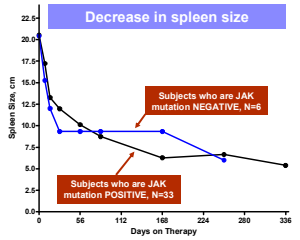
Subjects who are JAK mutation NEGATIVE, N=6
Subjects who are JAK mutation POSITIVE, N=33
Spleen Size, cm
Days on Therapy

Decrease in inflammatory cytokines: baseline vs Day 28

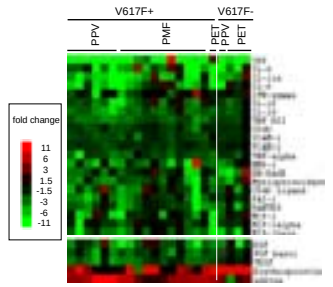
V617F+
V617F-
PPV
PMF
PET
PPV
PET
fold change
11
6
3
1.5
-1.5
-3
-6
-11

15 Summary and Conclusions

- Clinical correlates of V617F allele burden observed in study INCB18424-251 include
 - White cell counts
 - Spenomegaly
 - BMI
 - Plasma leptin
- INCB018424 therapy leads to a modest decrease in JAK2 V617F allele burden over time
 - Subset of patients have had more significant decreases in JAK2 V617F allele burden
- INCB018424 therapy results in significant clinical benefits independent of the magnitude of V617F allele burden decrease
- INCB018424 therapy results in significant clinical benefits in both JAK2 V617F-positive and JAK2 V617F-negative MF patients

16 References

- Antonioli E, Guglielmelli P, Poli G, et al. Influence of JAK2V617F allele burden on phenotype in essential thrombocythemia. *Haematologica*. 2008;93:41-8.
- Barosi G, Bergamaschi G, Marchetti M, et al. JAK2 V617F mutational status predicts progression to large splenomegaly and leukemic transformation in primary myelofibrosis. *Blood*. 2007;110:4030-6.
- Larsen TS, Pallisgaard N, Møller MB, Hasselbalch HC. The JAK2 V617F allele burden in essential thrombocythemia, polycythemia vera and primary myelofibrosis -- impact on disease phenotype. *Eur J Haematol*. 2007;79:508-15.
- Moliterno AR, Williams DM, Rogers O, Isaacs MA, Spivak JL. Phenotypic variability within the JAK2 V617F-positive MPD: roles of progenitor cell and neutrophil allele burdens. *Exp Hematol*. 2008;36:1480-6.
- Vannucchi AM, Antonioli E, Guglielmelli P, Pardanani A, Tefferi A. Clinical correlates of JAK2V617F presence or allele burden in myeloproliferative neoplasms: a critical reappraisal. *Leukemia*. 2008;22:1299-307.
- Wallace AM, Sattar N, McMillan DC. Effect of weight loss and the inflammatory response on leptin concentrations in gastrointestinal cancer patients. *Clin Cancer Res*. 1998;4:2977-9.

DISCLOSURES:

Drs. Verstovsek, Kantarjian, Pardanani, and Tefferi: *Have all acted in advisory and/or consulting capacities to Incyte Corp. and have received research funding from the Company.*

Drs. Burn, Vaddi, Redman, Bradley, Levy, Friedman, and Hollis: *Are all current or former employees of Incyte Corp. and have equity in the Company.*

THE DRIVE TO DISCOVER.
THE EXPERIENCE TO DELIVER.