The JAK Inhibitor INCB018424 Demonstrates Durable and Marked Clinical Responses in Primary Myelofibrosis (PMF) and Post-Polycythemia/Essential Thrombocythemia Myelofibrosis (Post-PV/ET-MF)

THE UNIVERSITY OF TEXAS MD ANDERSON CANCER CENTER Making Cancer History®

S. Verstovsek,[1] H. M. Kantarjian,[1] A. D. Pardanani,[2] D Thomas,[1] J. Cortes,[1] R. A. Mesa,[2] W. J. Hogan,[2] J. R. Redman,[3] S. Erickson-Viitanen,[3] R. Levy,[3], K. Vaddi,[3], E. Bradley,[3] J. Fridman,[3] and A. Tefferi[2]

[1]*University of Texas M. D. Anderson Cancer Center, Houston, Texas,* [2]*Mayo Clinic, Rochester, Minnesota, and* [3]*Incyte Corporation, Wilmington, Delaware, USA*

1 Abstract

Background: Myelofibrosis (MF) is characterized by progressive bone marrow dysfunction, extramedullary hematopoiesis, massive splenomegaly, elevated levels of inflammatory cytokines, severe constitutional symptoms, cachexia and premature death. INCB018424 is a potent, selective inhibitor of JAK 1 and 2 which has demonstrated impressive clinical activity in the early portion of a phase 1/2 study in patients with PMF and Post-PV/ET MF.

Methods: Following the initial dose escalation phase with a BID schedule (N=11), both twice daily (10 mg BID to 25 mg BID, N=56) and once daily (25 mg QD to 200 mg QD, N= 37) dose regimens were explored. Endpoints included spleen size assessed by manual palpation, constitutional symptoms assessed using the Modified Myelofibrosis Symptom Assessment Form (MFSAF) and JAK2 allele burden. Exercise capacity is being assessed using the Six Minute Walk Test (6MWT), and body mass and composition are being assessed using standard methods. Safety was assessed by collection of adverse events and monitoring of laboratory data.

Results: Over 100 patients are currently enrolled in the study, with a mean exposure to drug of > 5 months. Twenty percent of patients have received INCB018424 for > 9 months. INCB018424 is associated with a rapid reduction of splenomegaly in over 93% of patients from a mean baseline spleen size >20 cm, with 50% or greater reduction being observed in 35% of patients dosed with 10 mg BID or 50 mg QD, and 59% of patients dosed with 25mg BID regimens. These declines nearly always occur within the first month of therapy, with further, slower decreases often observed thereafter.

2 Abstract (*cont.*)

Results (*cont.*): In responding patients, spleen size reduction persists for the duration of therapy (currently up to one year follow-up). Constitutional symptoms are reported with high frequency in the study population as assessed by the MFSAF: night sweats, pruritis and fatigue were reported by >60%, >45% and >90% of patients respectively. Improvements in self-assessed symptom scores (mean % improvement of 40% to 60%) occurred by the first assessment at week 2. Activity limitations are also reported by a high proportion of MF patients; 70% of respondents reported impaired ability to exercise or get around, with a mean score >5 on the 11-point MFSAF scale; INCB018424 therapy was associated with rapid (within 2 weeks) and marked (40% or more) improvement in self-assessed score. There was a dose-dependent weight gain, most pronounced in patients with the lowest body mass index (BMI) values at baseline. Profound reductions in inflammatory cytokines were observed by the first evaluation at week 2 in virtually all patients, and were maintained during therapy. INCB018424 is generally well tolerated with the primary toxicity being dose-dependent grade 3 or 4 reversible thrombocytopenia which occurred in 0%, 18% and 32% of patients dosed with 10 mg BID, 50 mg QD or 25 mg BID.

Conclusions: INCB18424 shows unprecedented and durable clinical efficacy in reducing spleen size and improving constitutional symptoms in patients with MF. Results from proactive dose modification regimens, assessments of exercise capacity, MRI-based spleen volume assessment and body composition determination will be presented.

3 Hyperactivation of JAK/STAT signaling is pathogenic in MPDs

- An activating mutation in the pseudokinase domain of JAK2 known as V617F is present in
 - > 95% of polycythemia vera (PV)
 - ~ 50% of essential thrombocythemia (ET)
 - ~ 50% of primary myelofibrosis (PMF)
- Additional mutations that result in hyperactivation of JAK signaling include
 - Exon 12 mutations in JAK2
 - Mpl (TPO receptor) mutations – W515L, etc.
- JAK2/mpl mutations alone are causal in producing MPD phenotype in mice
- V617F allele burden in myelofibrosis correlates with
 - Poor survival
 - Increased leukemic transformation
 - Constitutional symptoms

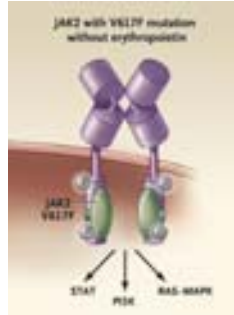

STAT
PI3K
RAS-MAPK

Source: *NEJM.* 2006:355:2452.

4 INCB018424

- Potent and selective ATP competitive JAK1 and JAK2 inhibitor
 - > 80-fold selective against JAK3 and panel of non-JAK kinases
- Inhibits signaling through mutant JAK2 and MPL pathways
 - Highly active in cellular assays driven by JAK2 V617
 - IC50 = 127 nM
 - Selective inhibition of V617F-driven EEC colony formation
- Inhibits pro-inflammatory cytokine expression and signaling in MF
 - Down-modulates cytokines and fibrogenic/angiogenic growth factors that play a potentially causal role in the pathogenesis of myelofibrosis
 - Inhibits the signaling of cytokines such as IL-6 which are known to be responsible for debilitating constitutional symptoms such as systemic inflammatory state and weight loss

5 INCB018424 Mechanism of Action

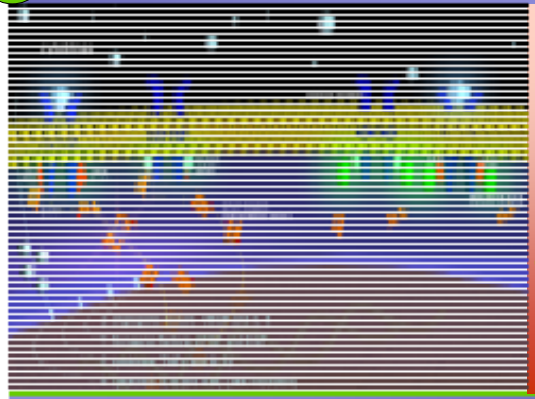

- INCB018424, a JAK1/2 inhibitor, blocks cytokine signaling and amplified cytokine and growth factor expression

6 INCB18424-251: Phase I/II Study of INCB18424 in Myelofibrosis

- Phase I dose escalation study of INCB018424 in PMF, post-PV MF and post-ET MF (ASH 2007)
 - Identified the starting dose of 25 mg BID as a highly effective dose in reducing splenomegaly and constitutional symptoms
 - Identified 25 mg BID as maximum tolerated dose (MTD), with reversible thrombocytopenia as the dose-limiting toxicity

- Expansion Cohort:
 - Safety and durability of efficacy upon long-term dosing
 - Evaluation of additional doses
 - Lower BID doses – 10 and 15 mg BID
 - Once daily doses – 25, 50, 100 and 200 mg
 - Evaluation of spleen size reduction by Magnetic Resonance Imaging
 - Prospective evaluation of improvement in constitutional symptoms associated with MF
 - Evaluation of functional end points of clinical efficacy
 - Overall daily activity

7 INCB18424-251: Patient Characteristics

Parameter	50 QD	10 mg BID	15 mg BID	25 mg BID
N	22	25	20	47
Median age (range)	65.5 (56-80)	58 (50-79)	65 (40-79)	67 (42-80)
Male/female ratio	10/12	15/10	13/7	31/16
PMF Post-PVMF Post-ETMF	14 (63.6%) 4 (18.2%) 4 (18.2%)	9 (36.0%) 12 (48.0%) 4 (16.0%)	12 (60.0%) 6 (30.0%) 2 (10.0%)	22 (46.8%) 17 (36.2%) 8 (17.0%)
Median time on drug	7.3 months	6.7 months	2.4 months	9.7 months
Percentage with JAK2 mutation	63.6%	95.7%	95.0%%	84.8%
Baseline spleen/liver size, cm Median (N with splenomegaly)	20.2 (N=21)	20.2 (N=23)	17.2 (N=20)	19.6 (N=39)
Baseline platelet ount, K/µL (mean ± SEM)	359 ± 272	333 ± 200	429 ± 186	345 ± 253
Percentage transfusion dependent	36%	32%	15%	38%

Includes only dose groups with at least 20 patients.

8 Study INCB18424-251: Disposition of Patients

Dose	N	Remain on study	Discontinued: Lack of response	Discontinued: Progression or toxicity	Discontinued: Other/ unrelated
10 mg BID	25	18	2	4	1
15 mg BID	20	20	0	0	0
25 mg BID	47	39	0	6	2
50 mg BID	5	2	0	2	1
25 mg QD	6	2	0	1	3
50 mg QD	22	18	1	1	2
100 mg QD	6	6	0	0	0
200 mg QD	3	3	0	0	0

- Most patients (108/134 patients) remain on study with a median duration on drug of 6.8 months

9 INCB18424-251: Adverse Events of at Least Moderate Intensity, All Causality

Adverse event	10 mg BID (N = 23)	15 mg BID (N = 20)	25 mg BID (N = 47)	50 mg QD (N = 22)
	Number (%) of subjects reporting			
Arthralgia	1 (4.3)	2 (10.0)	0	0
Diarrhea	1 (4.3)	0	1 (2.1)	1 (4.5)
Fatigue	2 (8.7)	0	1 (2.1)	0
Peripheral edema	1 (4.3)	0	2 (4.3)	1 (4.5)
Pyrexia	0	0	2 (4.3)	1 (4.5)
Pneumonia	0	0	1 (2.1)	2 (9.1)
Dyspnea	0	0	2 (4.3)	2 (9.1)

- Reported by at least 2% of the patients enrolled

10 INCB18424-251: Blood Counts

CTCAE grade	10 mg BID Hgb* (N = 15)	10 mg BID Platelets (N = 25)	15 mg BID Hgb* (N = 17)	15 mg BID Platelets (N = 20)	25 mg BID Hgb* (N = 29)	25 mg BID Platelets (N = 47)	50 mg QD Hgb* (N = 14)	50 mg QD Platelets (N = 22)
3	5 (33%)	1 (4.0%)	1 (5.9%)	0 –	7 (24%)	11 (23.4%)	7 (50%)	3 (13.6%)
4	1 (6.7%)	0 –	0 –	0 –	1 (3.4%)	3 (6.4%)	0 –	1 (4.5%)

*Includes only subjects who were transfusion independent at entry.

11 INCB18424-251: Safety Summary

- INCB018424 is generally well tolerated, with few adverse events other than mechanism-based effects on hematopoiesis
 - 108/134 patients remain on study with a median duration of ~7 months
- Reductions in platelet and Hgb counts are readily reversible with dose interruption and/or dose reduction while patients maintain clinical improvement
- Three transfusion-dependent MF patients in the study have achieved durable transfusion independence on INCB018424 therapy: one for > 1 year and 2 for > 6 months

12 INCB018424 Treatment Reduces Elevated Leukocytes and CD34+ Progenitor Cells

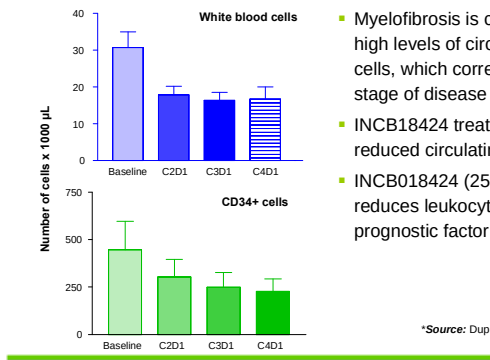

White blood cells
CD34+ cells
Number of cells x 1000 µL
Baseline
C2D1
C3D1
C4D1

- Myelofibrosis is characterized by high levels of circulating CD34+ cells, which correlate with the stage of disease
- INCB18424 treatment resulted in reduced circulating CD34+ cells
- INCB018424 (25 mg BID) reduces leukocytosis, an adverse prognostic factor, in MF patients*

*___Source:___ Dupriez et al. *Blood.* 1996;88:1013.

13 INCB018424 Treatment Results in Marked and Durable Improvement in Splenomegaly

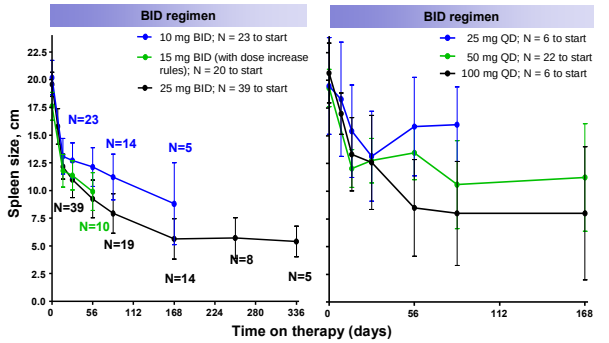

BID regimen
10 mg BID; N = 23 to start
15 mg BID (with dose increase rules); N = 20 to start
25 mg BID; N = 39 to start
BID regimen
25 mg QD; N = 6 to start
50 mg QD; N = 22 to start
100 mg QD; N = 6 to start
Spleen size, cm
N=23
N=14
N=5
N=39
N=10
N=19
N=14
N=8
N=5
Time on therapy (days)

Note: Data are censored after a dose change.

14 INCB018424 Treatment Results in Marked Increase in Overall Daily Activity Level

- Overall daily activity in 9 subjects was assessed using an accelerometer (pedometer) worn for 1 week periods of time at pre-dose and post 1 month dosing at 15 mg BID INCB018424
- Data reflects the difference in the average steps/day after 1 month therapy compared to baseline

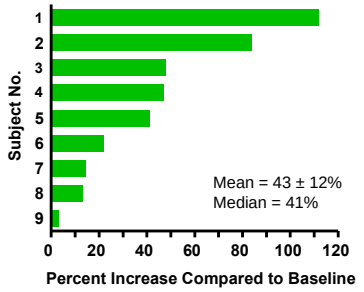

Subject No.
Mean = 43 ± 12%
Median = 41%
Percent Increase Compared to Baseline

Note: Data exclude 1 subject with severe pulmonary disease

14 INCB018424 Improves Splenomegaly Independent of JAK2 Mutation Status or Diagnosis

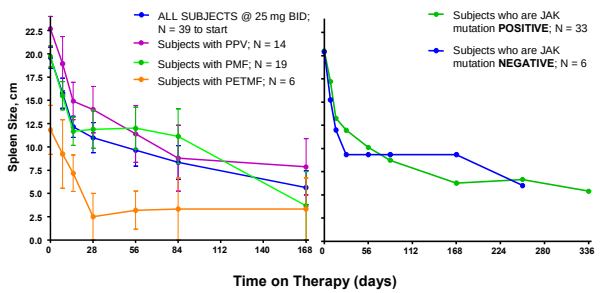

ALL SUBJECTS @ 25 mg BID; N = 39 to start
Subjects with PPV; N = 14
Subjects with PMF; N = 19
Subjects with PETMF; N = 6
Subjects who are JAK mutation POSITIVE; N = 33
Subjects who are JAK mutation NEGATIVE; N = 6
Spleen Size, cm
Time on Therapy (days)

Note: Data are censored after a dose change.

15 Objective Measurement by MRI Mirrors Spleen Size Reduction by Palpation After INCB018424 Treatment

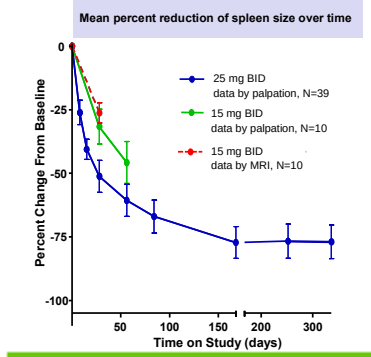

Mean percent reduction of spleen size over time
25 mg BID data by palpation, N=39
15 mg BID data by palpation, N=10
15 mg BID data by MRI, N=10
Percent Change From Baseline
Time on Study (days)

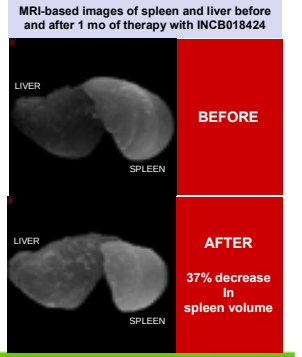

MRI-based images of spleen and liver before and after 1 mo of therapy with INCB018424
LIVER
SPLEEN
BEFORE
AFTER
37% decrease In spleen volume

17 INCB018424 Treatment Results in Rapid Improvement in Constitutional Symptoms

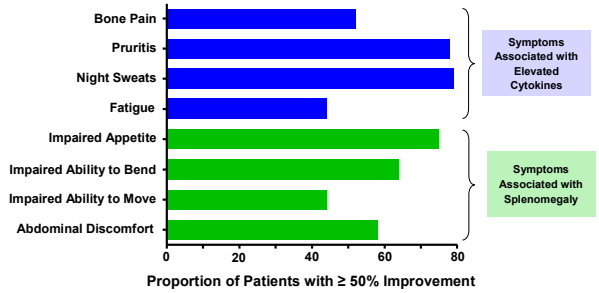

Bone Pain
Pruritis
Night Sweats
Fatigue
Impaired Appetite
Impaired Ability to Bend
Impaired Ability to Move
Abdominal Discomfort
Symptoms Associated with Elevated Cytokines
Symptoms Associated with Splenomegaly
Proportion of Patients with ≥ 50% Improvement

- Myelofibrosis Symptom Assessment Form (MFSAF) was used (N = 55 respondents)
- Improvements in symptoms were seen at the first on-treatment evaluation (2 weeks)

18 INCB18424-251: Summary

- INCB018424 treatment results in significant clinical benefits in myelofibrosis patients which include:
 - Decreased splenomegaly
 - Improvement in constitutional symptoms
 - Significant reduction in inflammatory cytokines (*ASH Abstract # 2804*)
 - Improvement of body weight (*ASH Abstract # 1760*)
 - Improvement of overall daily activity
- INCB18424 is very well tolerated
 - Median duration of therapy ~ 7 months
- INCB18424-251 results to date suggest that INCB018424 treatment results in significant clinical benefits to myelofibrosis patients, and represents a major advance as a therapy for this disease

DISCLOSURES:
Drs. Verstovsek, Kantarjian, Pardanani, Mesa, and Tefferi: *Have all acted in advisory and/or consulting capacities to Incyte Corp. and have received research funding from the Company.*
Drs. Redman, Erickson-Viitanen, Levy, Vaddi, Bradley, and Fridman: *Are all current or former employees of Incyte Corp. and have equity in the Company.*

Incyte THE DRIVE TO DISCOVER. THE EXPERIENCE TO DELIVER.