INCB018424, a Selective JAK1/2 Inhibitor, Significantly Improves the Compromised Nutritional Status and Frank Cachexia in Patients with Myelofibrosis (MF)

R. A. Mesa,[1] S. Friedman,[2] R. C. Newton,[2] S. Erickson-Viitanen,[2] S. Yeleswaram,[2] D. Hunter,[2] J. Redman,[2] S. Verstovsek,[3] H. M. Kantarjian,[3] A. D. Pardanani,[1] and A. Tefferi[1]

[1]*Mayo Clinic, Rochester, Minnesota,* [2]*Incyte Corporation, Wilmington, Delaware,* [3]*University of Texas M. D. Anderson Cancer Center, Houston, Texas, USA*

1 Abstract

Background: MF is a progressive and fatal illness associated with loss of lean muscle mass, cachexia, and weight loss (Mesa et al., *Cancer* 2007;109 (1):68-76) both from hypercatabolism (from increased pro-inflammatory cytokines) and MF associated splenomegaly, all of which are associated with decreased survival (Dupriez et al., *Blood* 1996;88:1013). Currently, there are no therapeutic interventions that have demonstrated a clinically significant impact on the progressive cachexia of MF. INCB018424 is a selective JAK1/2 inhibitor with the potential to improve the aberrant myeloproliferation in MF through decreasing signaling through constitutively active JAK-STAT activity, and nutritional status by decreasing splenomegaly and reducing pathologically increased cytokines.

Methods: Symptomatic patients enrolled in phase I/II trials (www.ClinicalTrials.gov, NCT00509899) with INCB018424 at the Mayo Clinic and at MD Anderson, were analyzed for the impact of therapy on nutritional status and cachexia. Specifically, in addition to traditional endpoints of the International Working Group for Myelofibrosis Research and Treatment (IWG-MRT) response (reported elsewhere), patients were assessed for changes in body mass index (BM = weight/(height * height)), serum cholesterol values, spleen size, and patient-reported assessment (by serial questionnaires) of early satiety and anorexia. In addition, leptin (another marker of nutritional state) is an adipose-derived protein hormone that plays a key role in regulating energy balance and circulates at levels proportional to body fat in health and disease (Mantovani et al, 2001; Fouladiun et al, 2005) was assessed at serial time points during therapy.

Results:

Patients: Thirty-four patients with MF, treated for at least 2 months with INCB018424 25 mg BID, were included in this analysis. Amongst this group 85% demonstrated splenomegaly (median 20 cm below left costal margin [range 4 cm to 32 cm], 6% with prior splenectomy) and had a median BMI of 24.8 (range 17.9 to 49.7). Although the median BMI at baseline is considered to be in the "normal range" (18.5-24.9), weight at baseline may well already reflect disease-related weight loss preceding enrollment and underestimate loss of lean muscle mass by artificial weight increases secondary to splenomegaly or edema.

Appetite: At enrollment, a clearly positive correlation between presence of anorexia and early satiety (by patient's report) and significant splenomegaly was observed. Treatment with INCB018424 led to resolution of symptoms of poor appetite, and early satiety, along with reduction in intra-abdominal organomegaly.

Weight: MF patients on therapy initially lost weight, which appears to reflect resolution of excess extravascular water (based on investigator-reported decreases in peripheral edema, ascites, or splenomegaly). As the trial progressed, MF patients on INCB018424 treatment progressively gained weight (mean increase of 0.40 kg, 2.93 kg, and 3.70 kg at 1, 2, and 3 months), both from improved appetite and potentially gain in lean muscle mass. Importantly, weight gain was more consistent, and of greater magnitude and durability, in patients who entered the study in the lowest quartile for BMI.

Cholesterol: We previously reported that hypocholesterolemia (total cholesterol < 150 or 100 mg/dl; HDL < 60 mg/dl) is associated with decreased survival in MF patients (Mesa et al., *Blood* 2007;110:a2548), potentially from hypercatabolism and splenic consumption of lipids. At enrollment, median total cholesterol was 95 mg/dl, with 94% and 55% below 150 mg/dl and 100 mg/dl, respectively. Following treatment with INCB018424, mean total cholesterol increased to 145 mg/dl (range 72-289 mg/dl) with 73% increasing to a improved range above their baseline (either to the > 100 mg/dl or > 150 mg/dl range).

Leptin: At enrollment, MF patients had very low leptin levels (mean = 2.55 ng/ml with 50% below 1 ng/ml vs. a range of 6-12 ng/ml for normal volunteers). Low plasma levels are associated with shortened survival in cancer patients. The plasma leptin levels increased 176% on average after 1 month of treatment with INCB018424, and continued to increase to levels matching healthy volunteers with time on study (mean = 7.04 ng/ml [range 0.25 – 35 ng/ml] after 2 months on INCB018424) and correlated to weight increases.

Conclusions: Therapy with INCB018424 improves the nutritional status of MF patients, including improving pathologic weight loss, hypercatabolism associated hypocholesterolemia, and pathologically decreased serum leptin. The improved nutritional status of MF patients treated with INCB018424 may reflect the ability of JAK inhibition to target the underlying pathophysiology of MF cachexia by reducing the organomegaly, levels of pro-inflammatory cytokines, and pro-inflammatory cytokine signaling intrinsic to the disease.

2 Background

- Myelofibrosis (MF) is a progressive illness associated with cachexia and weight loss*
 - These clinical signs, resulting from both hypercatabolism (secondary to increased pro-inflammatory cytokines) and MF-associated splenomegaly, are associated with decreased survival†
- Currently, there is no therapy that decreases the progressive cachexia of MF
- INCB018424 is a selective JAK1/2 inhibitor which has the potential to improve
 - the aberrant myeloproliferation in MF through decreasing constitutively active JAK-STAT signaling, and
 - nutritional status by decreasing both splenomegaly and the expression and signaling of pathologically increased cytokines.

**Source:* Mesa et al., *Cancer* 2007;109: 68. †*Source:* Dupriez et al., *Blood* 1996;88:1013.

3 INCB018424 Mechanism of Action

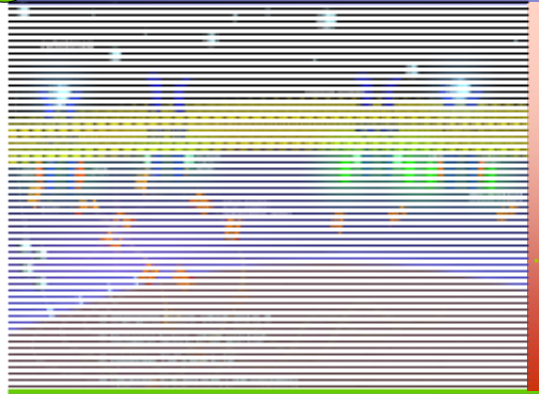
INCB018424, a JAK1/2 inhibitor, blocks cytokine signaling and amplified cytokine and growth factor expression

4 Study Design / Methods

- Symptomatic patients with MF enrolled in a phase I/II trial with INCB018424 (on the 25 mg BID cohort) were analyzed for the impact of therapy on nutritional status and cachexia*
- Patients were assessed for
 - Changes in body mass index (BMI)
 - Serum cholesterol values
 - Spleen size
 - Patient reports of early satiety and anorexia
- Leptin, an adipose-derived protein hormone that plays a key role in regulating energy balance and circulates at levels proportional to body fat in health and disease,† was assessed serially.

*For details of trial see www.ClinicalTrials.gov, NCT00509899.
†*Source:* Mantovani et al., *J Mol Med.* 2001;79: 406.

5 Patient Baseline Characteristics

Parameter	Value
N	47
Median age (range)	67.0 (42 to 80)
Male/female ratio	31/16
PMF Post-PVMF Post-ETMF	49% 33% 18%
Median time on drug	9.7 months
Percentage with JAK2 mutation	84.8%
Baseline spleen size, cm	20
Median no. patients with splenomegaly/hepatomegaly	8
Median BMI (range)	25.1 (17.9 to 49.7)

6 Summary of Results

- Patients
 - Thirty-four MF patients, treated for at least 2 months with INCB018424 25 mg BID, were included in this analysis (for baseline data see panel 5)
- Improvements in appetite
 - At enrollment, a clearly positive correlation between the presence of anorexia and early satiety (by patient's report) and significant splenomegaly was observed
 - Treatment with INCB018424 led to resolution of the symptoms of poor appetite and early satiety, along with the reduction in splenomegaly (see panels 8 and 9)
- Improvements in weight
 - MF patients on therapy initially lost weight, which reflects resolution of excess extravascular water (based on investigator-reported decreases in peripheral edema, ascites, or splenomegaly) (see panels 10 and 11).
 - As the trial progressed, MF patients on INCB018424 treatment progressively gained weight (mean increase of 0.40 kg at 1 month, 2.93 kg at 2 months, 3.70 at 3 months), and exhibited improved appetite
 - Weight gain was more consistent, of greater magnitude and more durable in patients who entered the study in the lowest quartile for BMI (see panel 12)

7 Summary of Results (*cont.*)

- Cholesterol
 - Hypocholesterolemia (total cholesterol < 150 or 100 mg/dl; HDL < 60 mg/dl) is associated with decreased survival in MF patients, potentially from hypercatabolism and splenic consumption of lipids (see panel 13)*
 - At enrollment, median total cholesterol was 95 mg/dl, with 94% and 55% below 150 mg/dl and 100 mg/dl, respectively.
 - Following treatment with INCB018424, median total cholesterol increased to 145 mg/dl (range 72-289 mg/dl) with 73% increasing to an improved range above their baseline (either to the > 100 mg/dl or the > 150 mg/dl range) (see panels 14 and 15).
- Leptin
 - At enrollment, MF patients had very low leptin levels (mean = 2.55 ng/ml with 50% below 1 ng/ml vs. a range of 6-12 ng/ml for normal elderly volunteers)
 - Cachetic MF patients have lower leptin levels than patients with GI cancers (see panel 16)
 - Low plasma levels are associated with shortened survival in cancer patients.
 - The plasma leptin levels increased 176% on average after 1 month of treatment with INCB018424; continued to increase to levels matching healthy volunteers with time on study (mean = 7.04 ng/ml (range 0.25 – 35 ng/ml) after 2 months on INCB018424; and correlated to weight increases (see panel 17).

**Source:* Mesa et al., *Cancer* 2007;109: 68.

8 Splenomegaly, Satiety, and Cachexia

- Splenomegaly is associated with symptoms of early satiety and cachexia

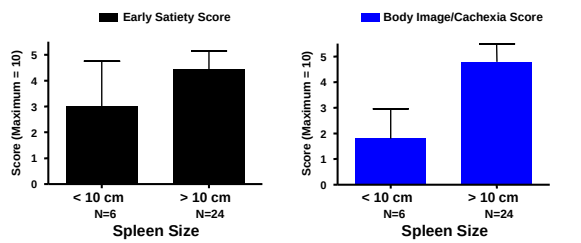
Early Satiety Score
Body Image/Cachexia Score
Score (Maximum = 10)
< 10 cm
N=6
> 10 cm
N=24
Spleen Size

9 INCB018424 and Symptoms of Splenomegaly

- Treatment with INCB018424 25 mg BID resolves symptoms of splenomegaly

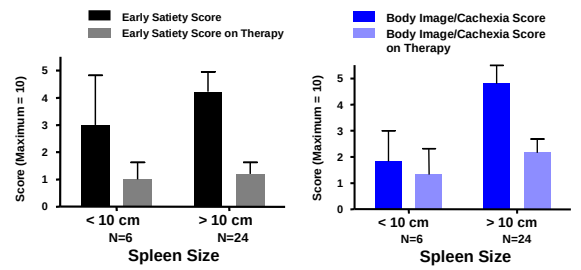
Early Satiety Score
Early Satiety Score on Therapy
Body Image/Cachexia Score
Body Image/Cachexia Score on Therapy
Score (Maximum = 10)
< 10 cm
N=6
> 10 cm
N=24
Spleen Size

10 Effect of INCB18424 on Body Weight

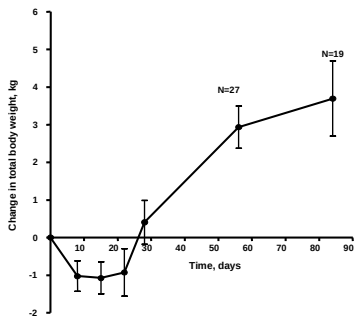
Change in total body weight, kg
Time, days
N=27
N=19

- Treatment with INCB018424 25 mg BID results in improved body weight in MF patients

11 Spleen Size and INCB18424 Treatment

- Spleen size reduction correlates with body weight gain following treatment with INCB018424

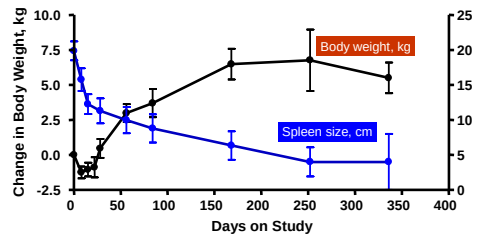
Change in Body Weight, kg
Spleen Size, cm
Days on Study
Body weight, kg
Spleen size, cm

Note: Includes all subjects (N=34) with censoring for dose change or dose interruption, but N values differ at each time point.

12 BMI and INCB18424 Treatment

- Patients in lowest BMI quartile gained more weight over more time than patients in highest BMI quartile

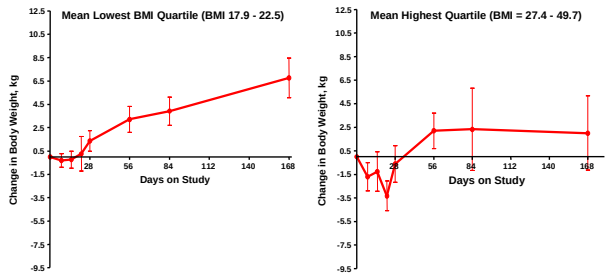
Mean Lowest BMI Quartile (BMI 17.9 - 22.5)
Mean Highest Quartile (BMI = 27.4 - 49.7)
Change in Body Weight, kg
Days on Study

13 INCB18424 and Serum Cholesterol

- Effect of INCB018424 treatment on total serum cholesterol level over time

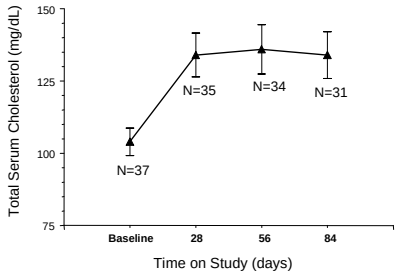
Total Serum Cholesterol (mg/dL)
N=37
N=35
N=34
N=31
Baseline
28
56
84
Time on Study (days)

14 Serum Cholesterol and Survival in MF

- Low serum cholesterol is associated with poor survival in patients with MF

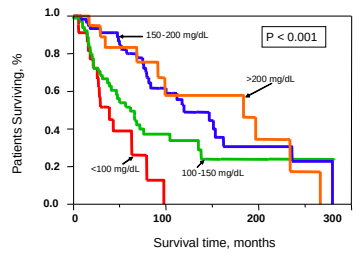
Patients Surviving, %
150-200 mg/dL
P < 0.001
>200 mg/dL
<100 mg/dL
100-150 mg/dL
Survival time, months

Reproduced by courtesy of Ruben Mesa, MD

15 INCB18424 and Total Cholesterol

- Effect of INCB018424 treatment on total serum cholesterol: categorical analysis

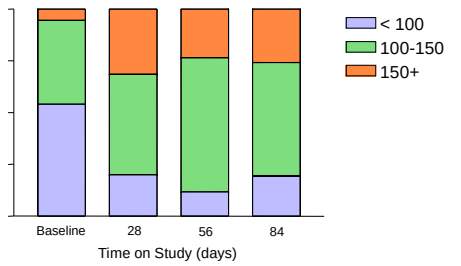
< 100
100-150
150+
Baseline
28
56
84
Time on Study (days)

16 Leptin Levels in Cachectic MF Patients

- Plasma leptin levels in cachectic MF patients are lower than those in elderly controls and GI cancer patients with cachexia

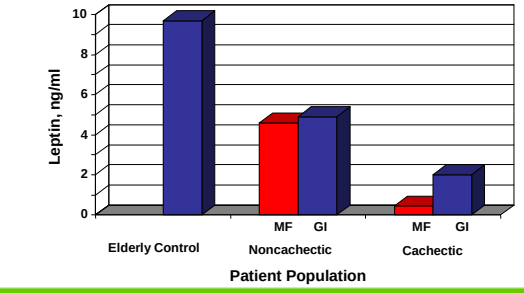
Leptin, ng/ml
Elderly Control
MF
GI
Noncachectic
Cachectic
Patient Population

17 INCB18424 Normalizes Leptin Levels

- INCB018424 raises abnormally low serum leptin levels into the normal range

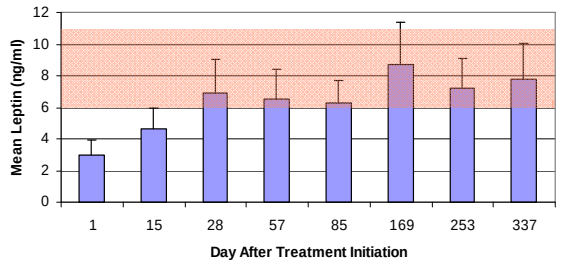
Mean Leptin (ng/ml)
Day After Treatment Initiation

18 Conclusions

- Therapy with INCB018424 improves the nutritional status of MF patients:
 - Pathologic weight loss
 - Hypercatabolism associated hypocholesterolemia
 - Pathologically decreased serum leptin
- Improved nutritional status of MF patients treated with INCB018424 may reflect ability of JAK inhibition to target underlying pathophysiology of MF cachexia by
 - Reducing organomegaly
 - Reducing levels of pro-inflammatory cytokines
 - Reducing pro-inflammatory cytokine signaling

DISCLOSURES:
Drs. Mesa, Verstovsek, Kantarjian, Pardanani, and Tefferi: *Have all acted in advisory and/or consulting capacities to Incyte Corp. and have received research funding from the Company.*
Drs. Friedman, Newton, Erickson-Viitanen, Redman, Yeleswaram, and Bradley: *Are all current or former employees of Incyte Corp. and have equity in the Company*

THE DRIVE TO DISCOVER.
THE EXPERIENCE TO DELIVER.