Preclinical characterization of INCB7839, a potent and selective inhibitor of ErbB ligand and HER2 receptor shedding: inhibition of ADAM10 and ADAM17 for the treatment of breast cancer

Jordan S Fridman, Peggy A Scherle, Xiangdong Liu, Eian Caulder, Michael Hansbury, Genjie Yang, Qian Wang, Yvonne Lo, Jincong Zhou, Wenqing Yao, Robert C Newton, Swamy Yeleswaram, Steven M Friedman, and Kris Vaddi

Incyte Corporation, Wilmington, Delaware, USA

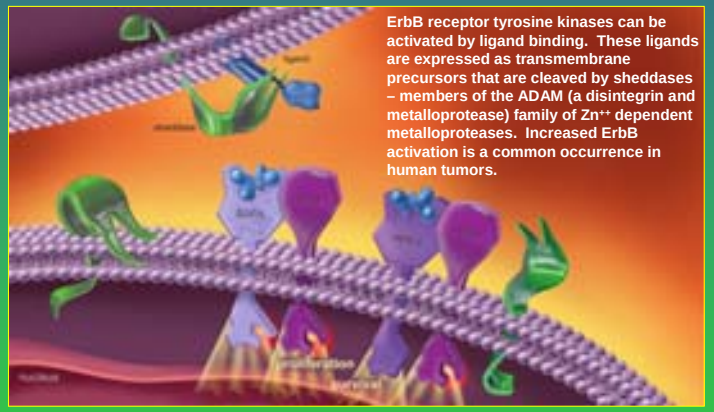
ErbB receptor tyrosine kinases can be activated by ligand binding. These ligands are expressed as transmembrane precursors that are cleaved by sheddases – members of the ADAM (a disintegrin and metalloprotease) family of Zn++ dependent metalloproteases. Increased ErbB activation is a common occurrence in human tumors.

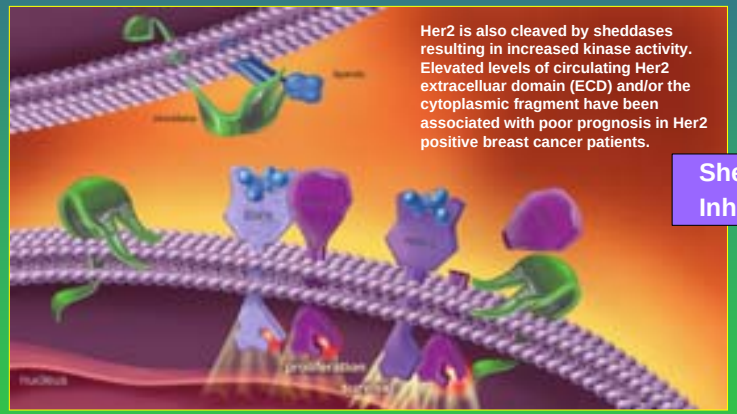
Her2 is also cleaved by sheddases resulting in increased kinase activity. Elevated levels of circulating Her2 extracellular domain (ECD) and/or the cytoplasmic fragment have been associated with poor prognosis in Her2 positive breast cancer patients.

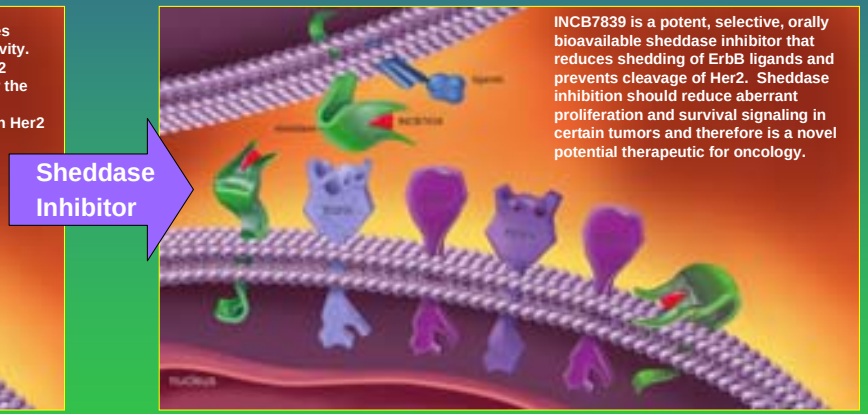
Sheddase Inhibitor
INCB7839 is a potent, selective, orally bioavailable sheddase inhibitor that reduces shedding of ErbB ligands and prevents cleavage of Her2. Sheddase inhibition should reduce aberrant proliferation and survival signaling in certain tumors and therefore is a novel potential therapeutic for oncology.

ABSTRACT

The ErbB family of receptor tyrosine kinases and the ligands that activate them are important regulators of cell proliferation, differentiation and survival. Dysregulation of this pathway occurs frequently in tumors of epithelial origin (including breast) and results in enhanced signal transduction from pathways associated with tumorigenesis and resistance to therapy. Interestingly, both the ligands that bind to the ErbB receptors (e.g. TGF and heregulin) and the HER2 receptor itself are proteolytically processed into active moieties. ErbB ligand cleavage generates a soluble, functionally active form that is critical for driving EGFR-mediated tumor cell growth and survival. Cleavage of the HER2 receptor results in the formation of a cytoplasmic fragment (p95) that possesses constitutive kinase activity and the release of an extracellular domain (ECD), which can be detected in circulation. Importantly, elevated levels of ECD and/or p95 have been associated with poor prognosis and both experimental and preliminary clinical data suggest that p95 affords resistance to trastuzumab. The enzymes responsible for these shedding events have been identified to be members of the ADAM family of metalloproteases, ADAM10 and ADAM17. Elevated ADAM activity has recently been shown to contribute to the transformed phenotype in various models of breast cancer and is associated with tumor progression. Hence, inhibition of these proteases represents a novel therapeutic strategy for the treatment of breast cancer. Toward this end, we have identified INCB7839, a potent, selective, orally bioavailable small molecule inhibitor of ADAM10 and ADAM17 which demonstrates selectivity against other metalloproteases. In xenograft models, INCB7839 effectively blocks HER2 cleavage in HER2 overexpressing human breast cancer cells in vitro and in vivo and dramatically enhances the antiproliferative effects of trastuzumab. INCB7839 also strikingly inhibits the growth of breast cancer cells lacking amplified HER2, through its effects on ErbB ligands, and potentiates the anti-tumor effects of a variety of cytotoxic, hormonal, and molecularly targeted anticancer agents. Mechanistic studies suggest that these effects are due, at least in part, to the ability of INCB7839 to reduce activation of the Akt survival pathway and increase the propensity of tumor cells to undergo apoptosis. In summary, inhibition of proteases responsible for activating ErbB pathways, through ligand and/or receptor cleavage, may offer a potentially novel and well tolerated therapeutic strategy for the treatment of human cancers. Toward testing this therapeutic hypothesis, INCB7839 is currently in clinical trials for the treatment of solid tumors.

1. INCB7839 is a potent, selective ADAM10/17 inhibitor (A) that reduces HER2 ECD shedding and p95 formation in cell lines (B) and *in vivo* (C)

A

Enzyme Assays		Marimastat IC_{50}, nM	INCB3619 IC_{50}, nM	INCB7839 IC_{50}, nM
ADAM9		254	>5000	>5000
ADAM10		78	37	19
ADAM17		12	17	43
MMP1		4	>5000	>5000
MMP2		3	35	131
MMP3		30	3500	>5000
MMP7		20	>5000	>5000
MMP9		<5	304	1106
MMP14		<5	772	1724

B

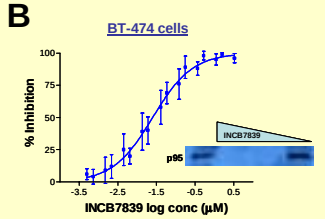
BT-474 cells
% Inhibition
INCB7839
p95
INCB7839 log conc (µM)

C

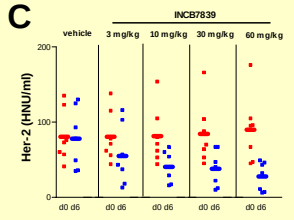
INCB7839
vehicle
3 mg/kg
10 mg/kg
30 mg/kg
60 mg/kg
Her-2 (HNU/ml)
d0 d6

2. INCB7839 potentiates the signaling and anti-proliferative effects of trastuzumab in HER2 shedding cells

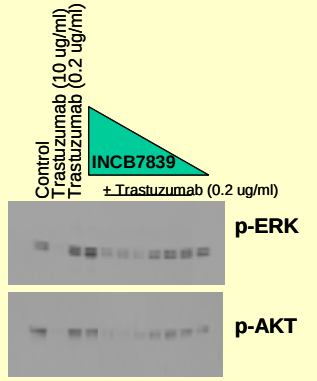
Control
Trastuzumab (10 ug/ml)
Trastuzumab (0.2 ug/ml)
INCB7839
+ Trastuzumab (0.2 ug/ml)
p-ERK
p-AKT

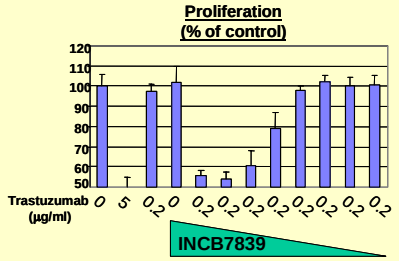
Proliferation (% of control)
Trastuzumab (µg/ml)
INCB7839

- •Similar results in SkBR3 cells
- •No potentiation observed in cells lacking amplified HER2
- •INCB7839 increases apoptosis induced by trastuzumab + taxane

3. Inhibition of HER2 shedding synergizes with of trastuzumab *in vivo* to control HER2+ disease

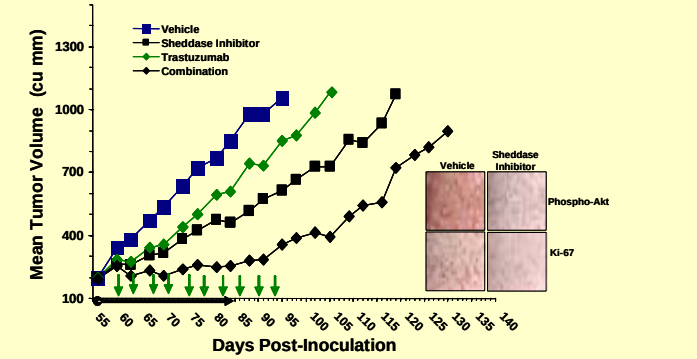
Vehicle
Sheddase Inhibitor
Trastuzumab
Combination
Mean Tumor Volume (cu mm)
Days Post-Inoculation
Vehicle
Sheddase Inhibitor
Phospho-Akt
Ki-67

*All mice received α-NK neutralizing antibodies to reduce ADCC

INCB7839 also potentiates the anti-tumor effects of lapatinib (Abstract #1117)

4. INCB7839 improves tumor growth control of a variety of classes of anti-cancer agents in HER2 amplified human tumor xenografts

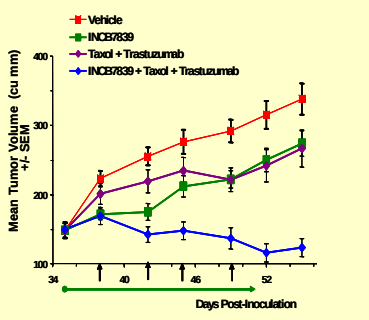
Vehicle
INCB7839
Taxol + Trastuzumab
INCB7839 + Taxol + Trastuzumab
Mean Tumor Volume (cu mm) +/- SEM
Days Post-Inoculation

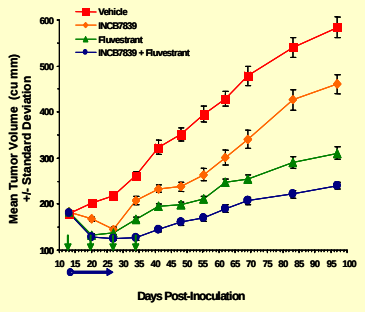
Vehicle
INCB7839
Fluvestrant
INCB7839 + Fluvestrant
Mean Tumor Volume (cu mm) +/- Standard Deviation
Days Post-Inoculation

5. Selective ADAM10/17 inhibition reduces ErbB ligand shedding *in vitro* (A & B) and *in vivo* (C)

A

PMA Stimulated Cell Lines

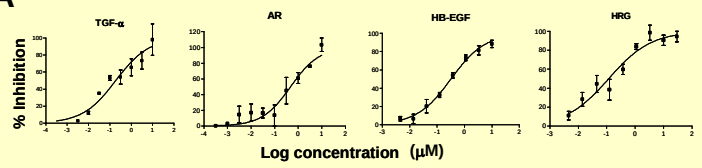
TGF-α
AR
HB-EGF
HRG
% Inhibition
Log concentration (µM)

Ectopic Expression of TGF-α

B

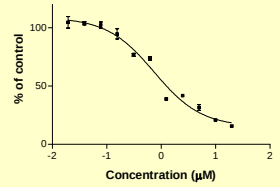
% of control
Concentration (µM)

C

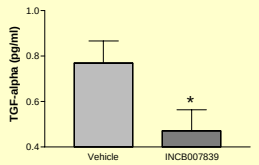
TGF-alpha (pg/ml)
Vehicle
INCB007839

6. INCB7839 curtails ErbB ligand dependent EGFR activation and tumor growth

Immortalized mouse mammary epithelial cells transformed with TGF-α

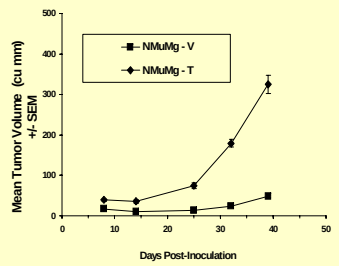
NMuMg - V
NMuMg - T
Mean Tumor Volume (cu mm) +/- SEM
Days Post-Inoculation

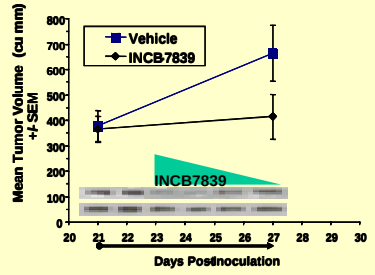
Vehicle
INCB7839
Mean Tumor Volume (cu mm) +/- SEM
Days Postinoculation

7. ADAM10/17 inhibition reduces ErbB mediated breast tumor xenograft growth (A) by impacting proliferation (B) and survival (B & C)

A

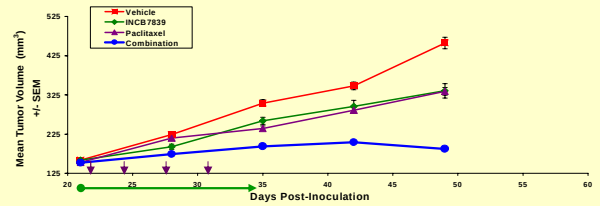
Vehicle
INCB7839
Paclitaxel
Combination
Mean Tumor Volume (mm³) +/- SEM
Days Post-Inoculation

B

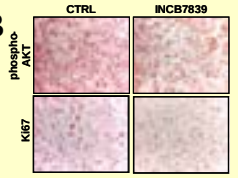
CTRL
INCB7839
phospho-AKT
Ki67

C

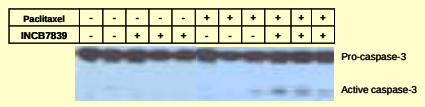
Paclitaxel
INCB7839
Pro-caspase-3
Active caspase-3

CONCLUSIONS

- A number of potent and selective ADAM10/17 inhibitors have been identified that reduce the shedding of Her2 ECD and ErbB ligands *in vitro* and *in vivo*.
- Incyte sheddase inhibitors demonstrate single agent activity and/or synergize with cytotoxic and targeted therapies in breast cancer xenograft tumor models deemed responsive to ErbB manipulation.
- INCB7839 was well tolerated at efficacious doses and does not induce the musculoskeletal side effects observed with broad-spectrum MMP inhibitors in multiple preclinical models (not shown).
- INCB7839 is currently being evaluated in the clinic (Abstract # 6064) .

The Drive to Discover. The Experience to Deliver.