A multicenter phase Ib study of the safety, pharmacokinetics, biological activity and clinical efficacy of INCB7839, a potent and selective inhibitor of ADAM10 and ADAM17

Jeffrey Infante, MD[1], Howard A Burris, MD[1], Nancy Lewis, MD[2], Ross Donehower, MD[3], John Redman, MD[4], Steven Friedman, MD[4], Peggy Scherle, PhD[4], Jordan Fridman, PhD[4], Jun Li, PhD[4], Thomas Emm, PhD[4], Steve Troy, MS[4] and S Gail Eckhardt, MD[5]. *[1]Sarah Cannon Research Institute, Nashville, TN, USA, 37203; [2]Fox Chase Cancer Center, Philadelphia, PA, USA, 19111; [3]Johns Hopkins Cancer Center, Baltimore, MD, USA, 21231; [4]Incyte Corporation, Wilmington, DE, USA, 19880 and [5]University of Colorado Health Sciences Center, Denver, CO, USA, 80045.*

Background

Signaling through the ErbB family of receptor tyrosine kinases regulates tumor cell proliferation and survival. Both the ligands that bind to the ErbB receptors as well as the HER2 receptor itself are proteolytically processed into biologically active moieties by members of the ADAM family of metalloproteases, specifically ADAM10 and ADAM17 ErbB ligand cleavage generates a soluble, functionally active form that is critical for driving EGFR-mediated tumor cell growth and survival. Elevated ErbB ligand levels have been associated with poor prognosis and can diminish the efficacy of TKIs directed against the ErbB family of receptors. Cleavage of the HER2 receptor results in the formation of a cytoplasmic fragment (p95) that possesses constitutive kinase activity and the release of an extracellular domain (ECD), which can be detected in circulation. Elevated levels of ECD and/or p95 have been associated with poor prognosis and preliminary data suggest that p95 affords resistance to trastuzumab.

We have identified INCB7839, a potent, selective, orally bioavailable first in class, small molecule inhibitor of ADAM10 and ADAM17 that has convenient oral dosing using capsules, high solubility with excellent oral bioavailability and sustained formulation successfully developed to prolong exposure and reduce peak (maintains metalloprotease selectivity profile). In preclinical models, INCB7839: effectively blocks HER2 cleavage in HER2 overexpressing human breast cancer cells and enhances the antiproliferative effects of HER2 targeted therapies, including trastuzumab and lapatinib; inhibits ErbB ligand cleavage and potentiates the anti-tumor effects of a variety of cytotoxic, hormonal, and targeted anticancer agents in cell lines lacking HER2 amplification; and was well-tolerated in 28-day safety studies in rat and cynomolgus monkey. ADAM inhibition therefore represents a potentially novel therapeutic strategy for the treatment of ErbB receptor family driven tumors.

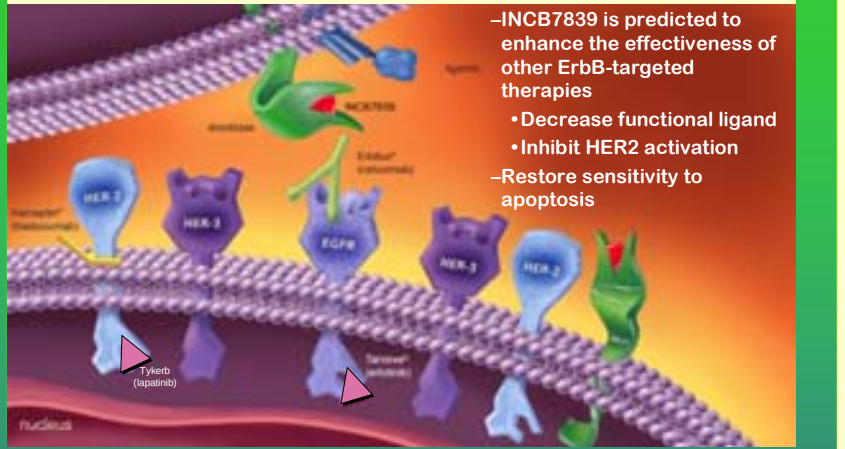
–INCB7839 is predicted to enhance the effectiveness of other ErbB-targeted therapies
•Decrease functional ligand
•Inhibit HER2 activation
–Restore sensitivity to apoptosis
HER-2
HER-3
EGFR
Tykerb (lapatinib)
nucleus

Objectives

- **Primary**: Establish MTD of INCB7839 slow release (SR) formulation given BID for 28 days
- **Secondary**:
 - Evaluate safety profile
 - Evaluate PK
 - Explore biological activity on EGFR pathway-related biomarkers:
 - Plasma HER2 ECD levels
 - Plasma EGFR ligand levels (Heregulin, Amphiregulin, TGFα)
 - Explore single agent efficacy in refractory patient populations

Enrollment Criteria

Inclusion

- 18 years or older with cancer refractory to standard treatment
 - non-small cell lung cancer
 - hormone refractory prostate cancer
 - colorectal cancer
 - head and neck squamous cell cancer
 - breast cancer
- Life expectancy of 12 weeks or longer.
- Able to comprehend and willing to sign an informed consent form
- ECOG performance status of 0 or 1
- Has ECG with a QTcii range < 440 msec for males and < 460 msec for females
- A negative screening serum pregnancy test for females of childbearing potential
- Males and female of childbearing potential agree to utilize pregnancy prevention techniques

Exclusion

- Received any anticancer medications or investigational study drug in the 28 days prior to receiving their first dose of study medication (hormonal treatments are allowed)
- Evidence of active hepatitis or HIV infection
- Brain metastases or spinal cord compression
- Significantly impaired renal or hepatic function or inadequate bone marrow reserve
- Surgery within 4 weeks prior to study entry, excluding the placement of vascular access
- Evidence of venous thrombosis by flow Doppler examination
- A history of thrombosis or a coagulation disorder. Patients with a history of DVT need to be on therapeutic anticoagulation
- Contraindication to the use of low dose warfarin and /or low dose aspirin

Study Design/Patient Demographics

- BID oral dosing regimen
- "3 + 3 design" (with fourth patient in a cohort optional)
- Toxicity was evaluated continuously and laboratory tests (hematology plasma d-dimer and serum chemistry) were performed weekly
- Flow Doppler scan of legs and abdomen every 2 weeks in the first cycle and at the start of each subsequent cycle
- Tumor evaluations were performed every 8 weeks for patients treated on the 28-day schedule
- DLT defined as any AE ≥ grade 3, at least possibly drug-related and clearly not related to disease progression
- If a DLT occurs in ≥ two patients in a cohort, then cohort dose one level below will be the MTD. At least six patients treated at MTD with no more than one patient with a DLT will confirm this as the recommended phase II dose.

Patient Demographics

Median Age	Years	63
	Range	(37 – 84)
Gender	Female	17
	Male	13
ECOG PS	0	14
	1	16
Cancer types	CRC	12
	Breast	10
	Prostate	6
	Head and Neck	2
Number of prior chemotherapy regimens	1-2	3
	3-4	14
	5-7	8
	>7	4

Study Enrollment by Dose Level

Cohort	Dose	Number of Evaluable Patients	Total Number of Dosed Patients
1	100 mg IR BID	5	7
2	200 mg IR BID	3	4
3	200 mg SR BID	5	7
4	300 mg SR BID	5	9
5	300 mg SR BID + warfarin	2	3

IR = immediate release formulation SR = sustained release formulation

Adverse Events

- Generally well tolerated
 - No significant EGFR TKI-related toxicities: no rashes and only occasional mild diarrhea
 - No MMP associated toxicities: no evidence of musculoskeletal syndrome (MSS)
 - No compound related alterations in clinical chemistries, including liver enzymes, or effects on formed elements of the blood
- Thrombotic Events
 - 5 of 27 patients developed lower extremity DVTs during the first 28 day treatment cycle
 - One additional patient with stable disease for 4 cycles experienced a PE
 - In the 300 mg SR cohort, 2 DLTs occurred (both DVTs) so MTD without supportive interventions is defined as 200 mg SR BID
- INCB7839 has no other identified toxicity

INCB7839 Pharmacokinetics

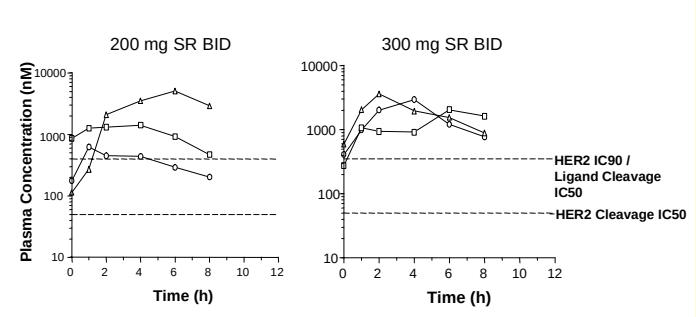
200 mg SR BID
300 mg SR BID
Plasma Concentration (nM)
Time (h)
HER2 IC90 / Ligand Cleavage IC50
HER2 Cleavage IC50

INCB7839 exposures at both 200mg and 300 mg SR doses are projected to significantly inhibit both HER2 and EGFR ligand cleavage

INCB7839 Significantly Inhibits HER2 Shedding

Serum HER2 ECD as a Biomarker in Breast Cancer

- 50-80% of patients with HER2 overexpressing breast cancer have elevated circulating levels of Her-2 extracellular domain (ECD)
- Elevated ECD predicts lower response to treatment and shortened survival
- Changes in serum HER2 ECD levels are associated with clinical outcome
- **Study 201 includes three HER2 overexpressing breast cancer patients with elevated HER2 ECD levels**

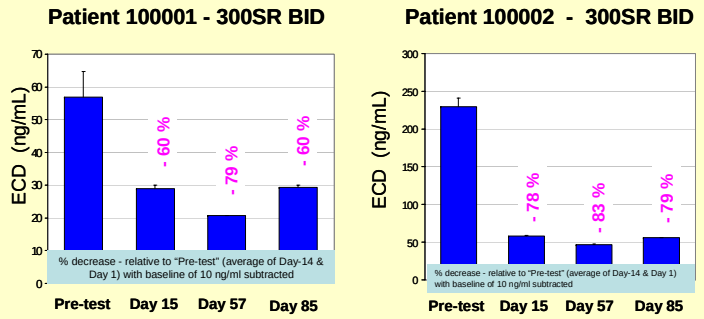
Patient 100001 - 300SR BID
Patient 100002 - 300SR BID
ECD (ng/mL)
- 60 %
- 79 %
- 60 %
- 78 %
- 83 %
- 79 %
% decrease - relative to "Pre-test" (average of Day-14 & Day 1) with baseline of 10 ng/ml subtracted
Pre-test
Day 15
Day 57
Day 85

Patient 30010, dosed with 200SR BID INCB7839 was not evaluated beyond day 7 due to disease progression but demonstrated a 51% decrease in plasma ECD by day 7

INCB7839 Significantly Inhibits EGFR Ligand Shedding

Serum EGFR Ligand Levels as a Biomarker

- Levels are low to undetectable in most cancer patients
- Increased levels of amphiregulin and TGF-a predict poor response to gefitinib in NSCLC
- **Study 201 includes two subjects with elevated plasma ErbB ligand levels**
 - Due to short plasma half life of EGFR ligands, effect of INCB7839 on ligand levels assessed on day 15 during PK sampling

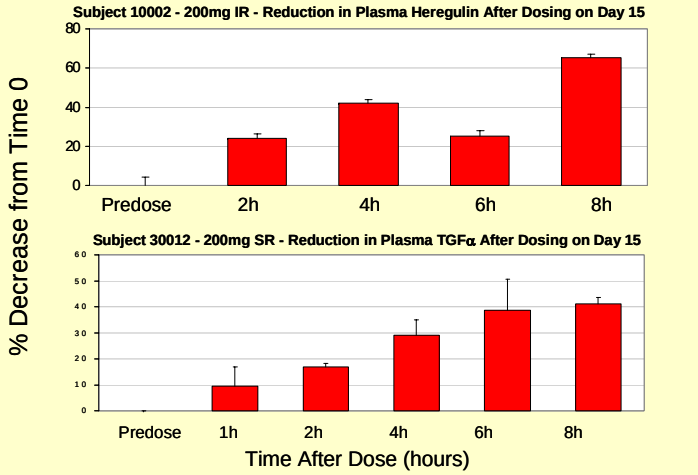
Subject 10002 - 200mg IR - Reduction in Plasma Heregulin After Dosing on Day 15
Subject 30012 - 200mg SR - Reduction in Plasma TGFα After Dosing on Day 15
% Decrease from Time 0
Predose
1h
2h
4h
6h
8h
Time After Dose (hours)

Antitumor Activity

- 30 subjects enrolled in 5 cohorts
 - 20 evaluable (completed at least 1 cycle)
- 6 subjects demonstrated stable disease after 2 months of therapy
- 6 evaluable subjects with HER2+ breast cancer
 - All had failed trastuzumab containing regimens
 - 4/6 patients demonstrated stable disease from 2-4 months including both evaluable patients with elevated ECD

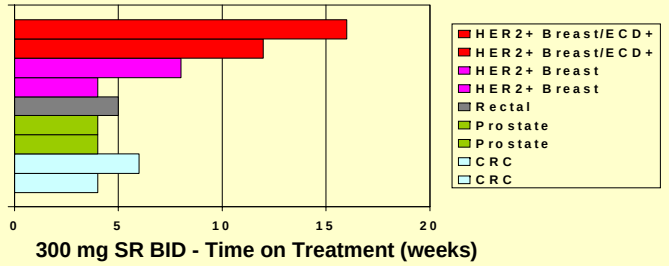
HER2+ Breast/ECD+
HER2+ Breast/ECD+
HER2+ Breast
HER2+ Breast
Rectal
Prostate
Prostate
CRC
CRC
300 mg SR BID - Time on Treatment (weeks)

CT Scan – Patient 100001 (300 SR BID)

January 07 March 07

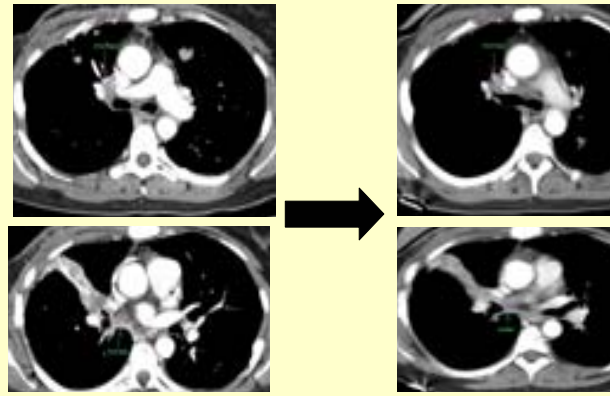

Conclusions

- Successful transition to SR formulation
- INCB7839 significantly impacts PD markers: inhibiting both HER2 ECD and EGFR ligand levels in cancer patients
- 6/20 evaluable subjects demonstrated stable disease including 4/7 patients administered the highest dose tested to date
- 4/6 patients with HER2+ breast cancer demonstrated stable disease including both evaluable patients with elevated ECD
- With exception of DVT no other identified toxicity at doses which impact the targeted biology and yield clinical efficacy
- DVT issue can be addressed
 - No subject who entered on prophylactic anticoagulation experienced a thrombotic event while on study

Study Status

- Dose escalation has restarted using INCB7839 coadministered with low dose warfarin and short dose holidays every cycle
- Future studies combining the MTD of INCB7839 with ErbB receptor family targeted therapies are planned

Incyte

The Drive to Discover. The Experience to Deliver.